Zürcher Hochschule
für Angewandte Wissenschaften
zh aw Life Sciences und Facility Management
A.Vogel

Optimization and validation of an automated sample preparation of a herbal drug on the basis of a Solid-Phase-Extraction

Murtaj, V.*; Krienbühl H.; Leng A.**; Meier B.***

***) Zurich University of Applied Science, 8820 Wädenswil Switzerland**
****) Bioforce AG, 9325 Roggwil, Switzerland**

Introduction

Herbal medicinal products contain one or several extracts, and each of them consists of a multitude of compounds. Quantitative analysis of analytical markers is therefore challenging. In addition, the matrix of the galenical formulation may affect the analysis. Therefore, extraction of specific markers during sample preparation is sometimes inevitable. However, it is time consuming and is a potential source for errors. Automation of sample preparation may help to reduce handling time and inconsistent data due to manual sample processing.

Objective of this study is to characterise a new automated SPE system (SmartPrep™). On the basis of the obtained results, an existing Liquid-liquid extraction LLE sample preparation method for an Echinacea preparation is to be replaced by the automated SPE system. The new method is validated according to the ICH guidelines for analytical validations.

To guarantee batch to batch uniformity of the active ingredient, efficient identification and quantification of the main substance (dodeca-2E,4E,8Z,10E/Z-tetraenoic acid isobutylamide) in a matrix-loaded Echinacea preparation needs to be ensured.

Material and Methods

Test solution
An Echinaforce® preparation was analysed. The preparation mainly consist a fluid extract of purple coneflower (Echinacea purpurea L.), high amounts of sucrose and a fruit concentrate. It is manufactured by Bioforce AG, CH-9325 Roggwil.

Reference substance
A standard solution provided by Bioforce AG with a concentration of 0.333 mg dodecatetraenoic acid isobutylamide per 100 ml was used as reference substance.

Equipment
Device for sample preparations: SmartPrep™ automated SPE-Extractor with Chromabond C18, 3 ml, 500 mg cartridge (Macherey-Nagel).

UPLC parameters
The content of dodecatetraenoic acetic acid isobutylamide in the Echinacea preparation was determined by means of a Waters ACQUITY UPLC ® System. The assay was carried out with a validated RP-UHPLC method using an alkylamide standard from Bioforce AG.

Column: Waters UPLC column, HSS T3, 1.8 µm, 100 mm x 2.1 mm.
Mobile phase: Gradient with acetonitrile and 0.5 % trifluoroacetic acid in water
Flow rate: 0,5 ml/min.
Injection volume: 8 µl.
Column temperature: 40 °C.
UV detection wavelength: 260 nm.
Running time: 8 min.

SPE steps of automation:

Table 1: Optimized and established SPE-sample preparation method with 3 ml C-18 SPE cartridges

SPE function	Reagent and volume	Sip rate [ml min^{-1}]	Delivery rate [ml min^{-1}]
Clean Plunger	3 ml 100 % Methanol	-	10
Clean Plunger	3 ml 40 % Methanol	-	10
Condition cartridge	3 ml 100 % Methanol	-	5
Condition cartridge	3 ml 40 % Methanol	-	5
Load sample	15 ml sample	10	5
Clean sample line	5 ml 40 % Methanol	-	10
Load sample	6 ml sample	10	5
Clean sample line	5 ml 40 % Methanol	-	10
Load sample	6 ml sample	10	5
Wash cartridge	2,5 ml 40 % Methanol	5	-
Wash cartridge	2,5 ml 40 % Methanol	5	-
Elute cartridge	7,5 ml 100 % Methanol	5	-
Clean sample line	3 ml 40 % Methanol	10	-

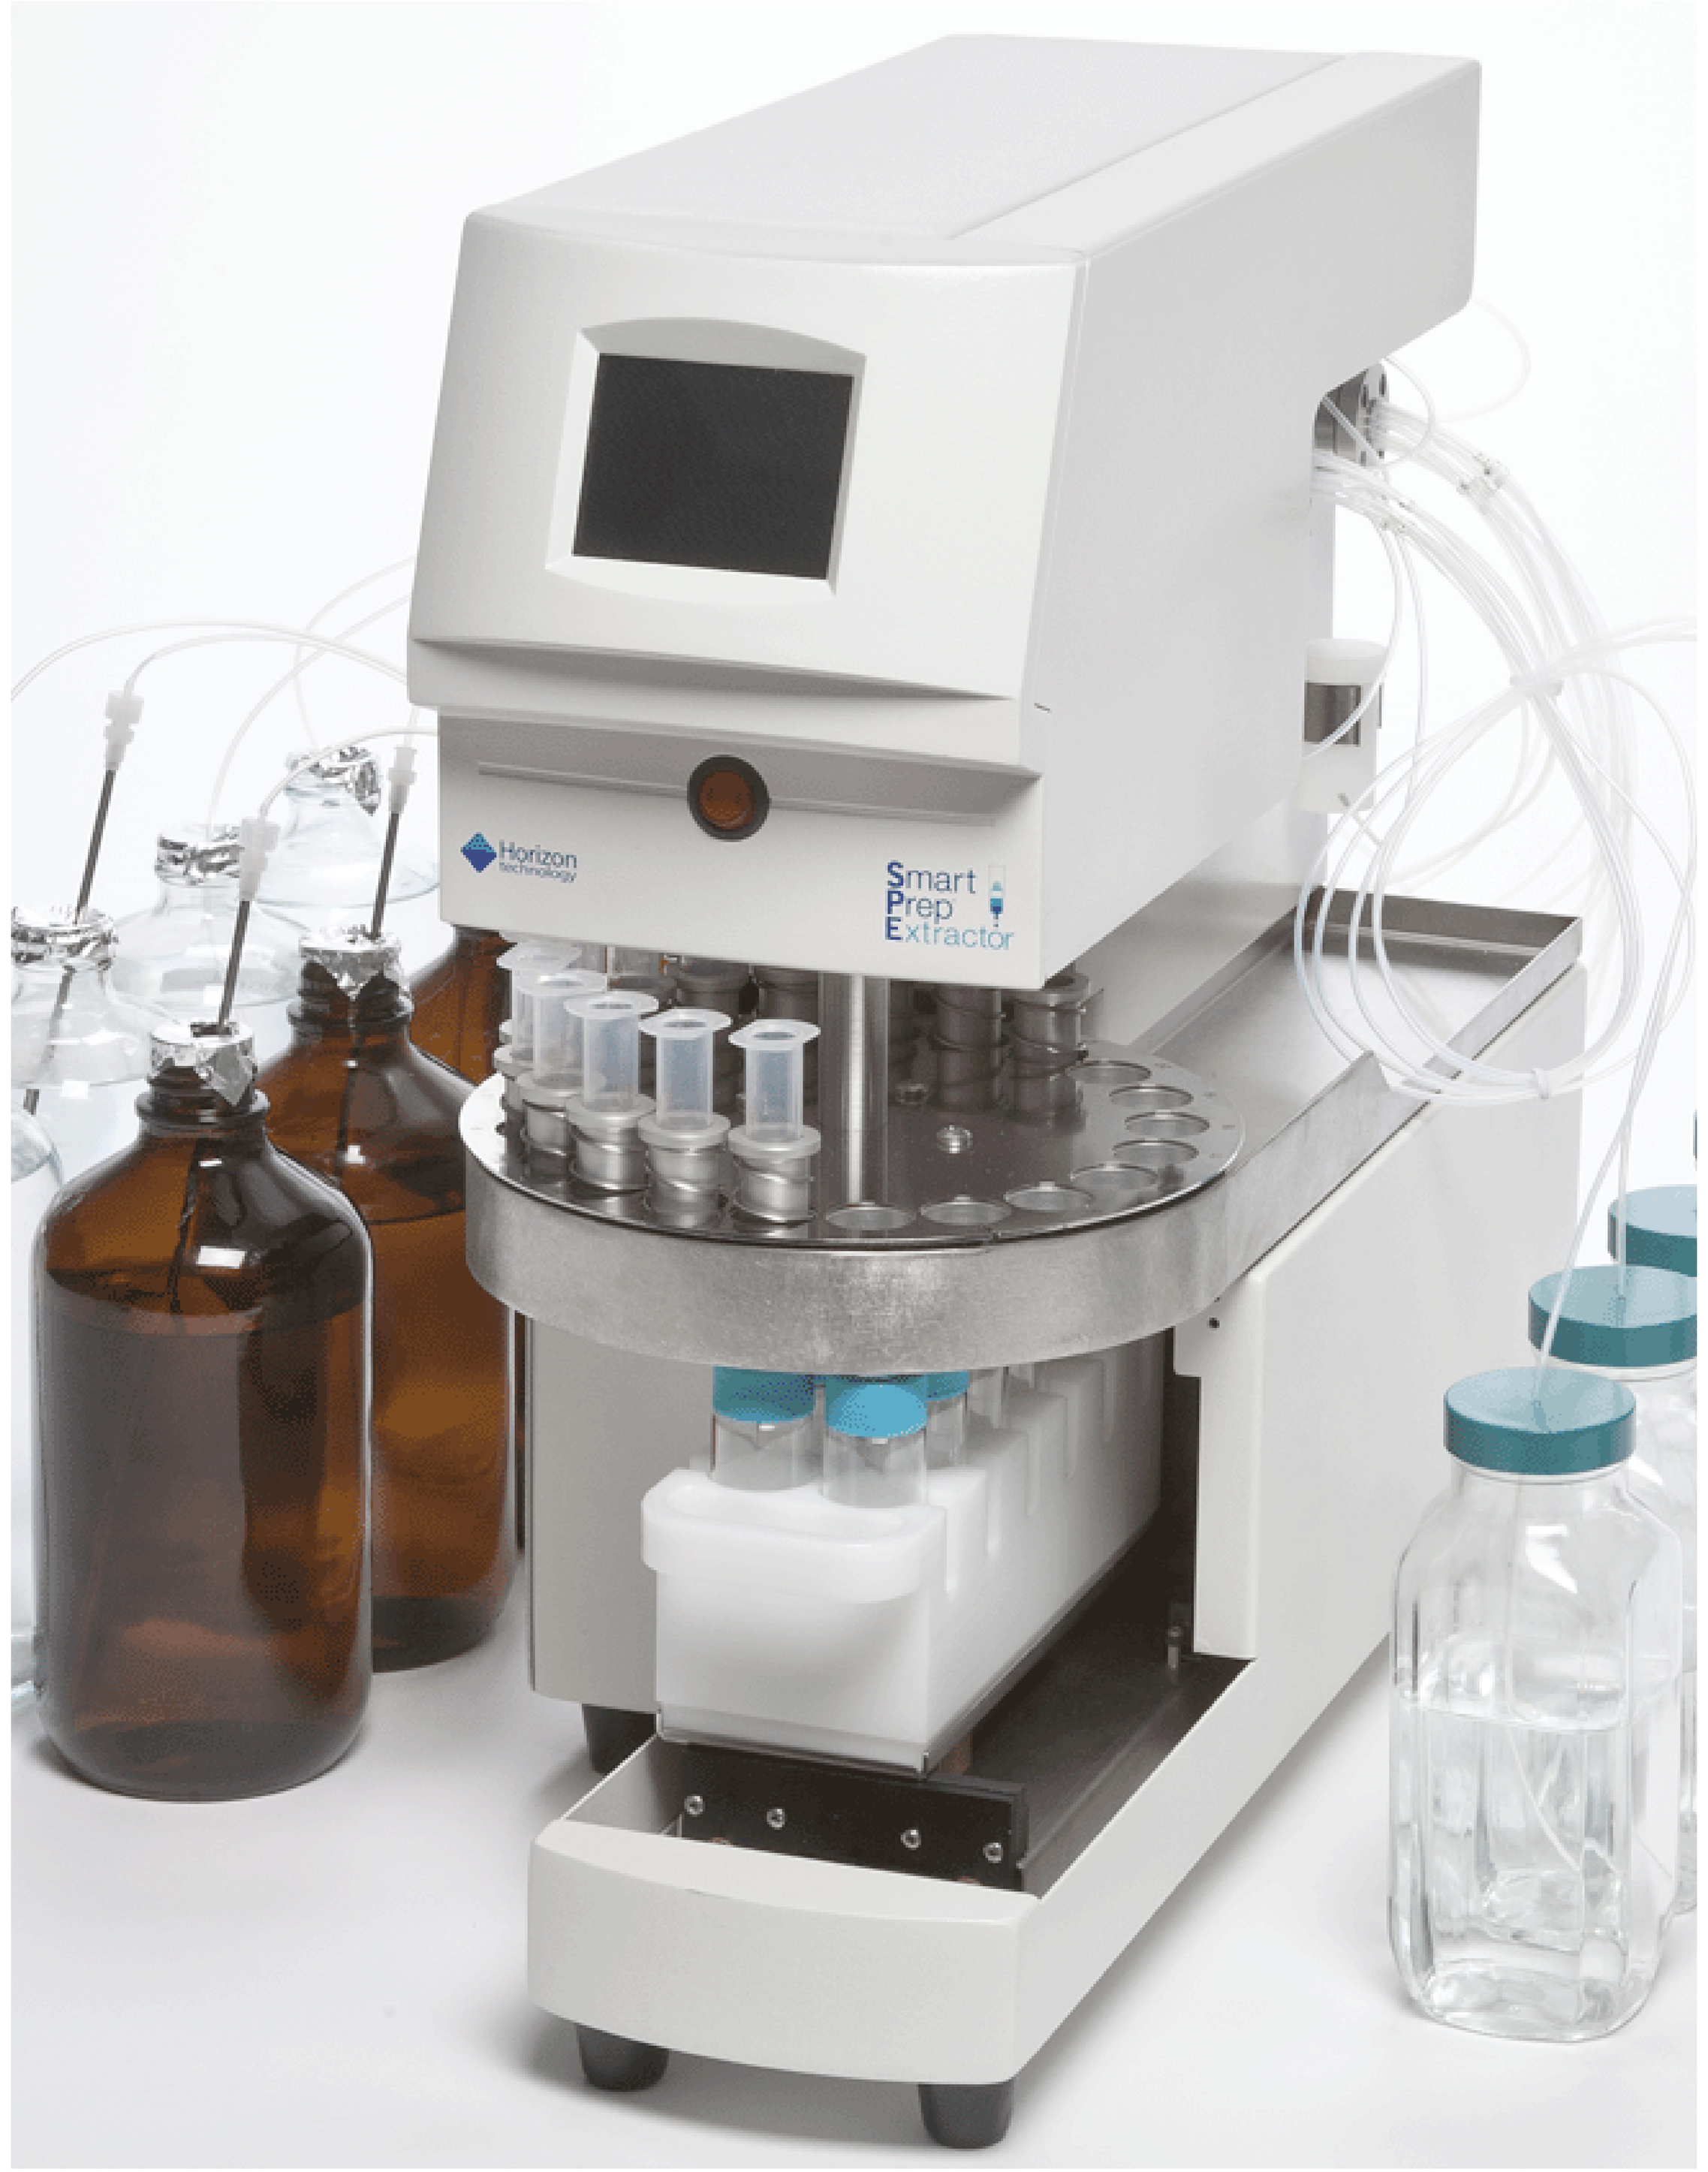
Horizon technology
Smart
Prep
Extractor

Figure 1: SmartPrep™ - Automated solid-phase extraction system by Horizon Technology for sample processing of *Echinacea* preparation

Results

Figure 2 shows the linearity of the automated SPE method over the whole specified range. The apparent linear range covers the amount of 0.1 g up to 2.5 g. Duplicate determinations on 7 concentration levels shows, based on the sample amount and peak area, the following regression: y =77940 x + 834.1 and R^2 0.99987.

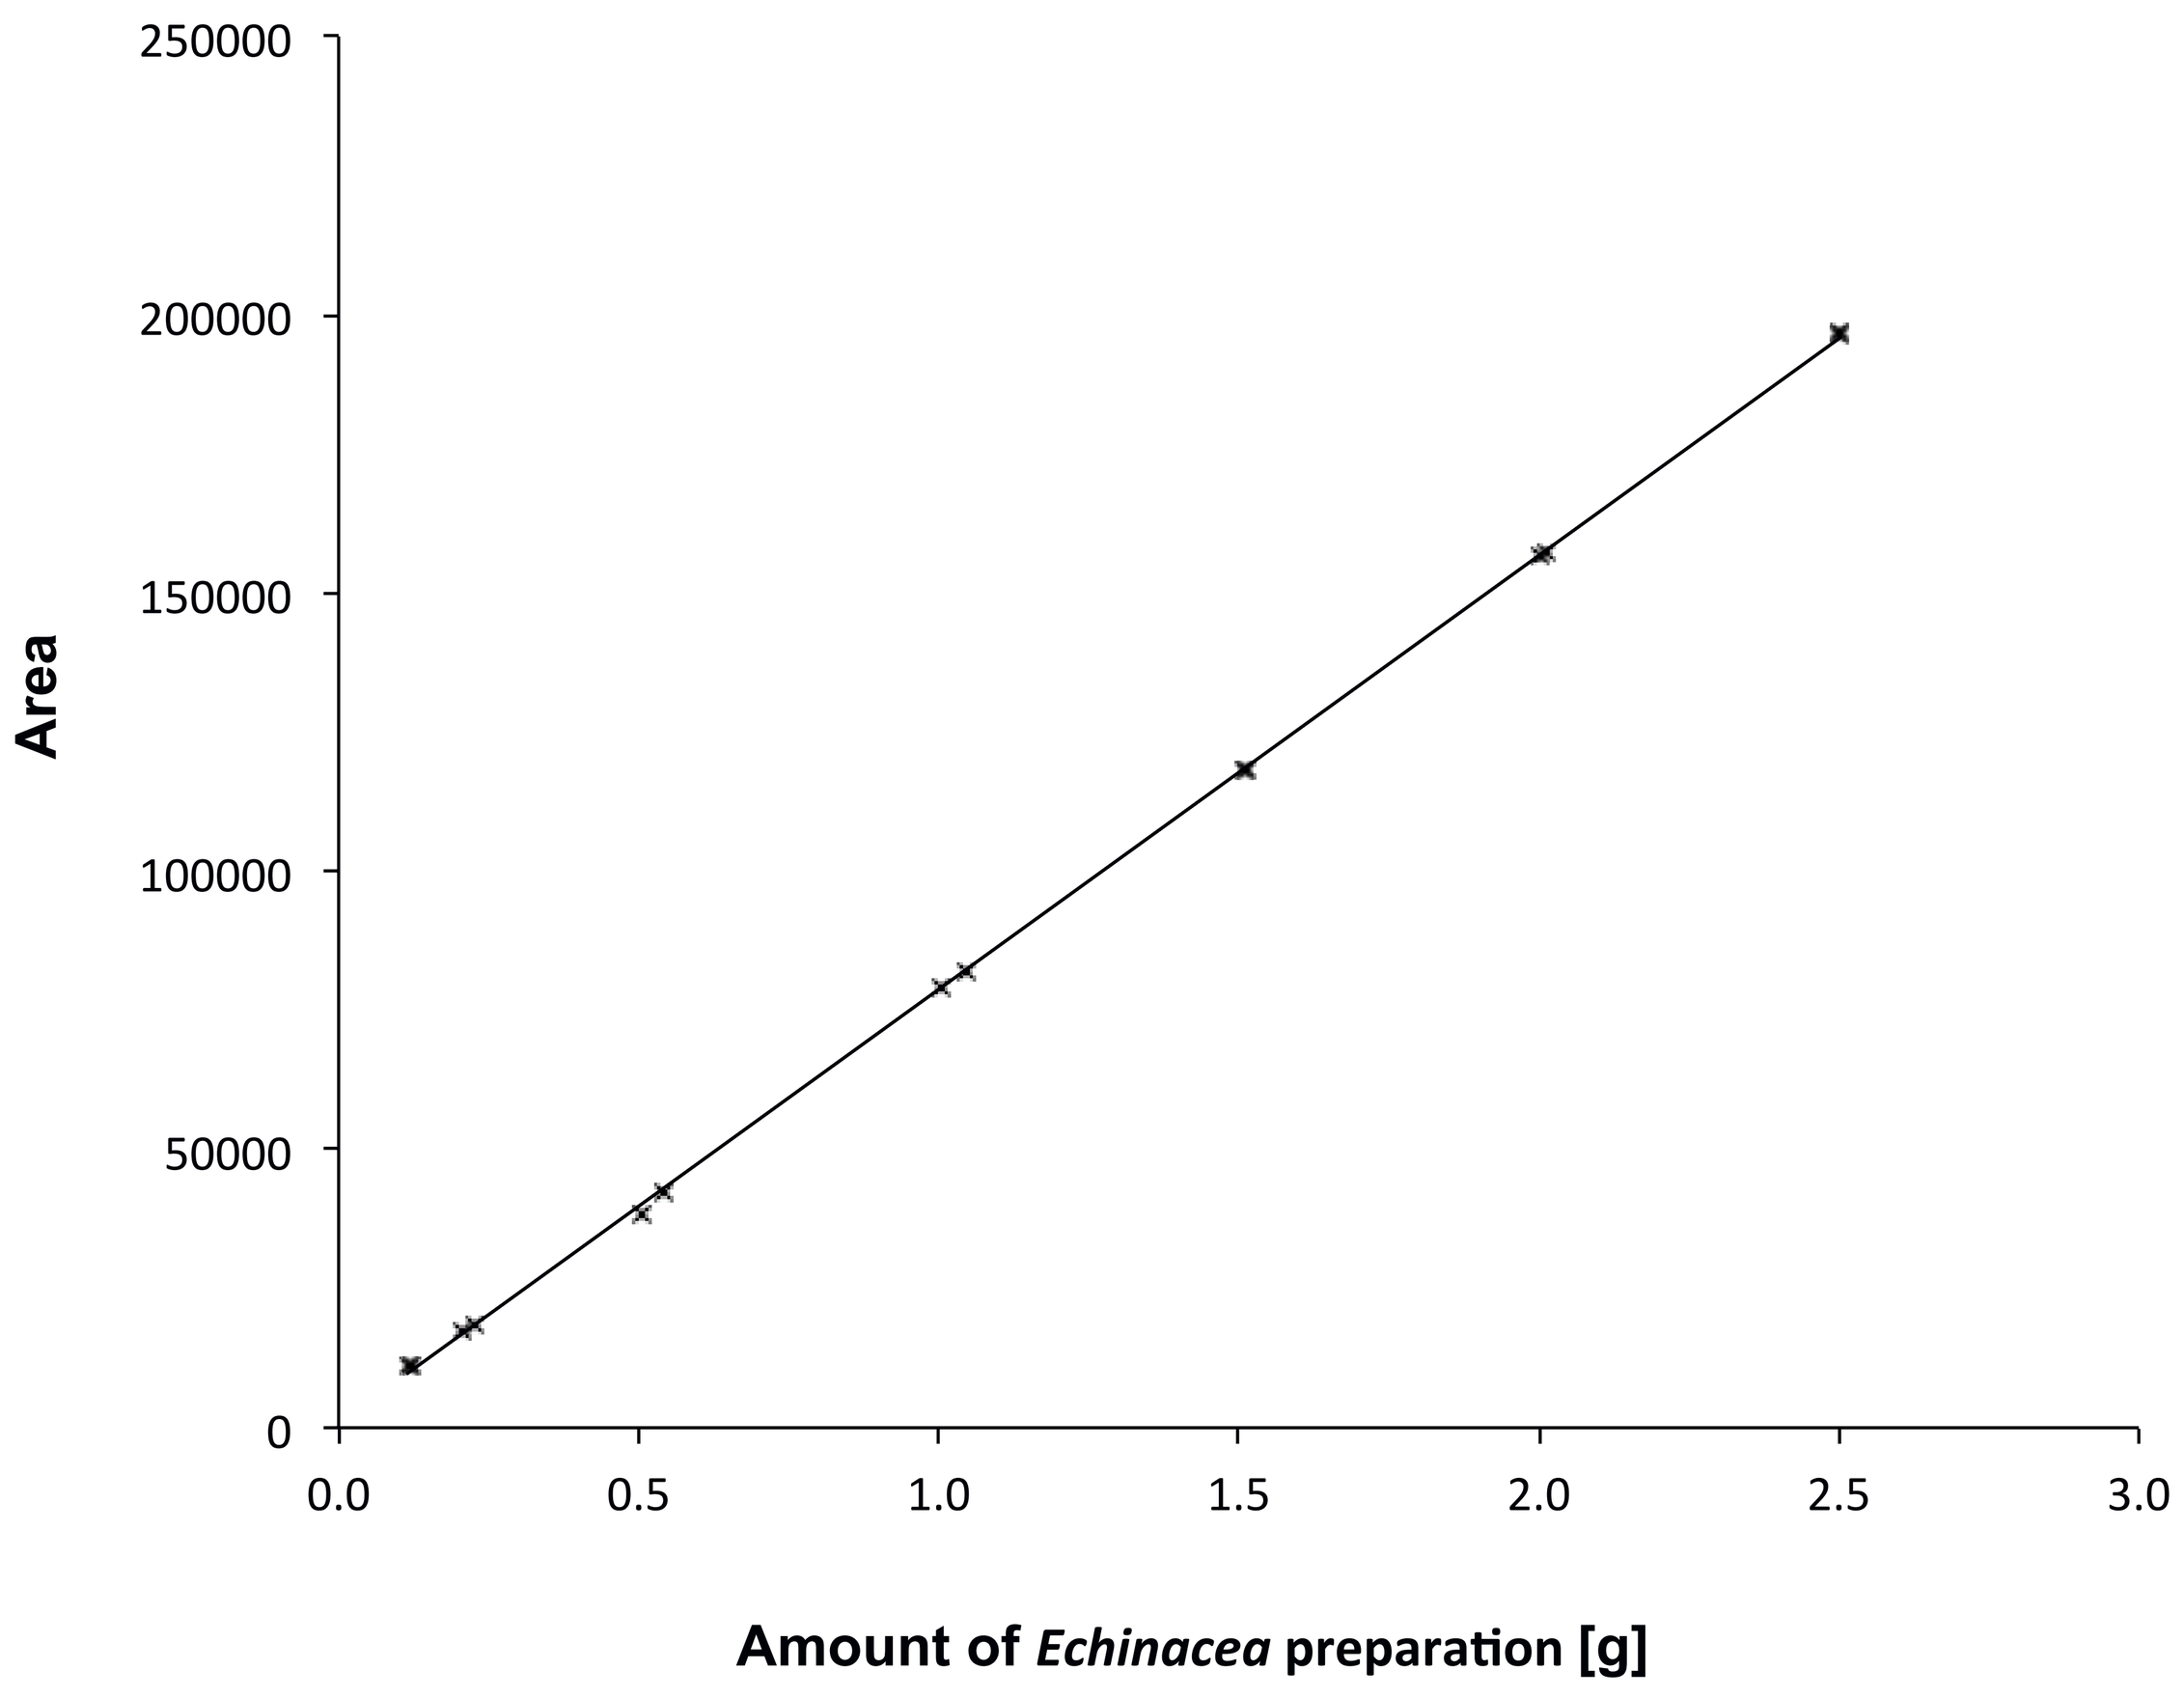
250000
200000
150000
100000
50000
0
Area
0.0
0.5
1.0
1.5
2.0
2.5
3.0
Amount of Echinacea preparation [g]

Figure 2: Verification of the linearity by comparing the peak area against the concentration of the Echinacea preparation using the final SPE-method on the SmartPrep™.

Figure 3 shows the yield of dodeca-2E,4E,8Z,10E/Z-tetraenoic acid isobutylamide when using the newly established SPE-method on the SmartPrep™ in comparison to the SPE-vacuum technique and the LLE-method (liquid-liquid extraction). The amount of 0.707 mg dodeca-2E,4E,8Z,10E/Z-tetraenoic acid isobutylamide per 100 g is achieved by SmartPrep™ method, generating a 4.1 % higher outcome than by the LLE-test procedure.

In comparison to the SPE vacuum technique, a 2.1 % higher concentration is achieved when using the SmartPrep™.

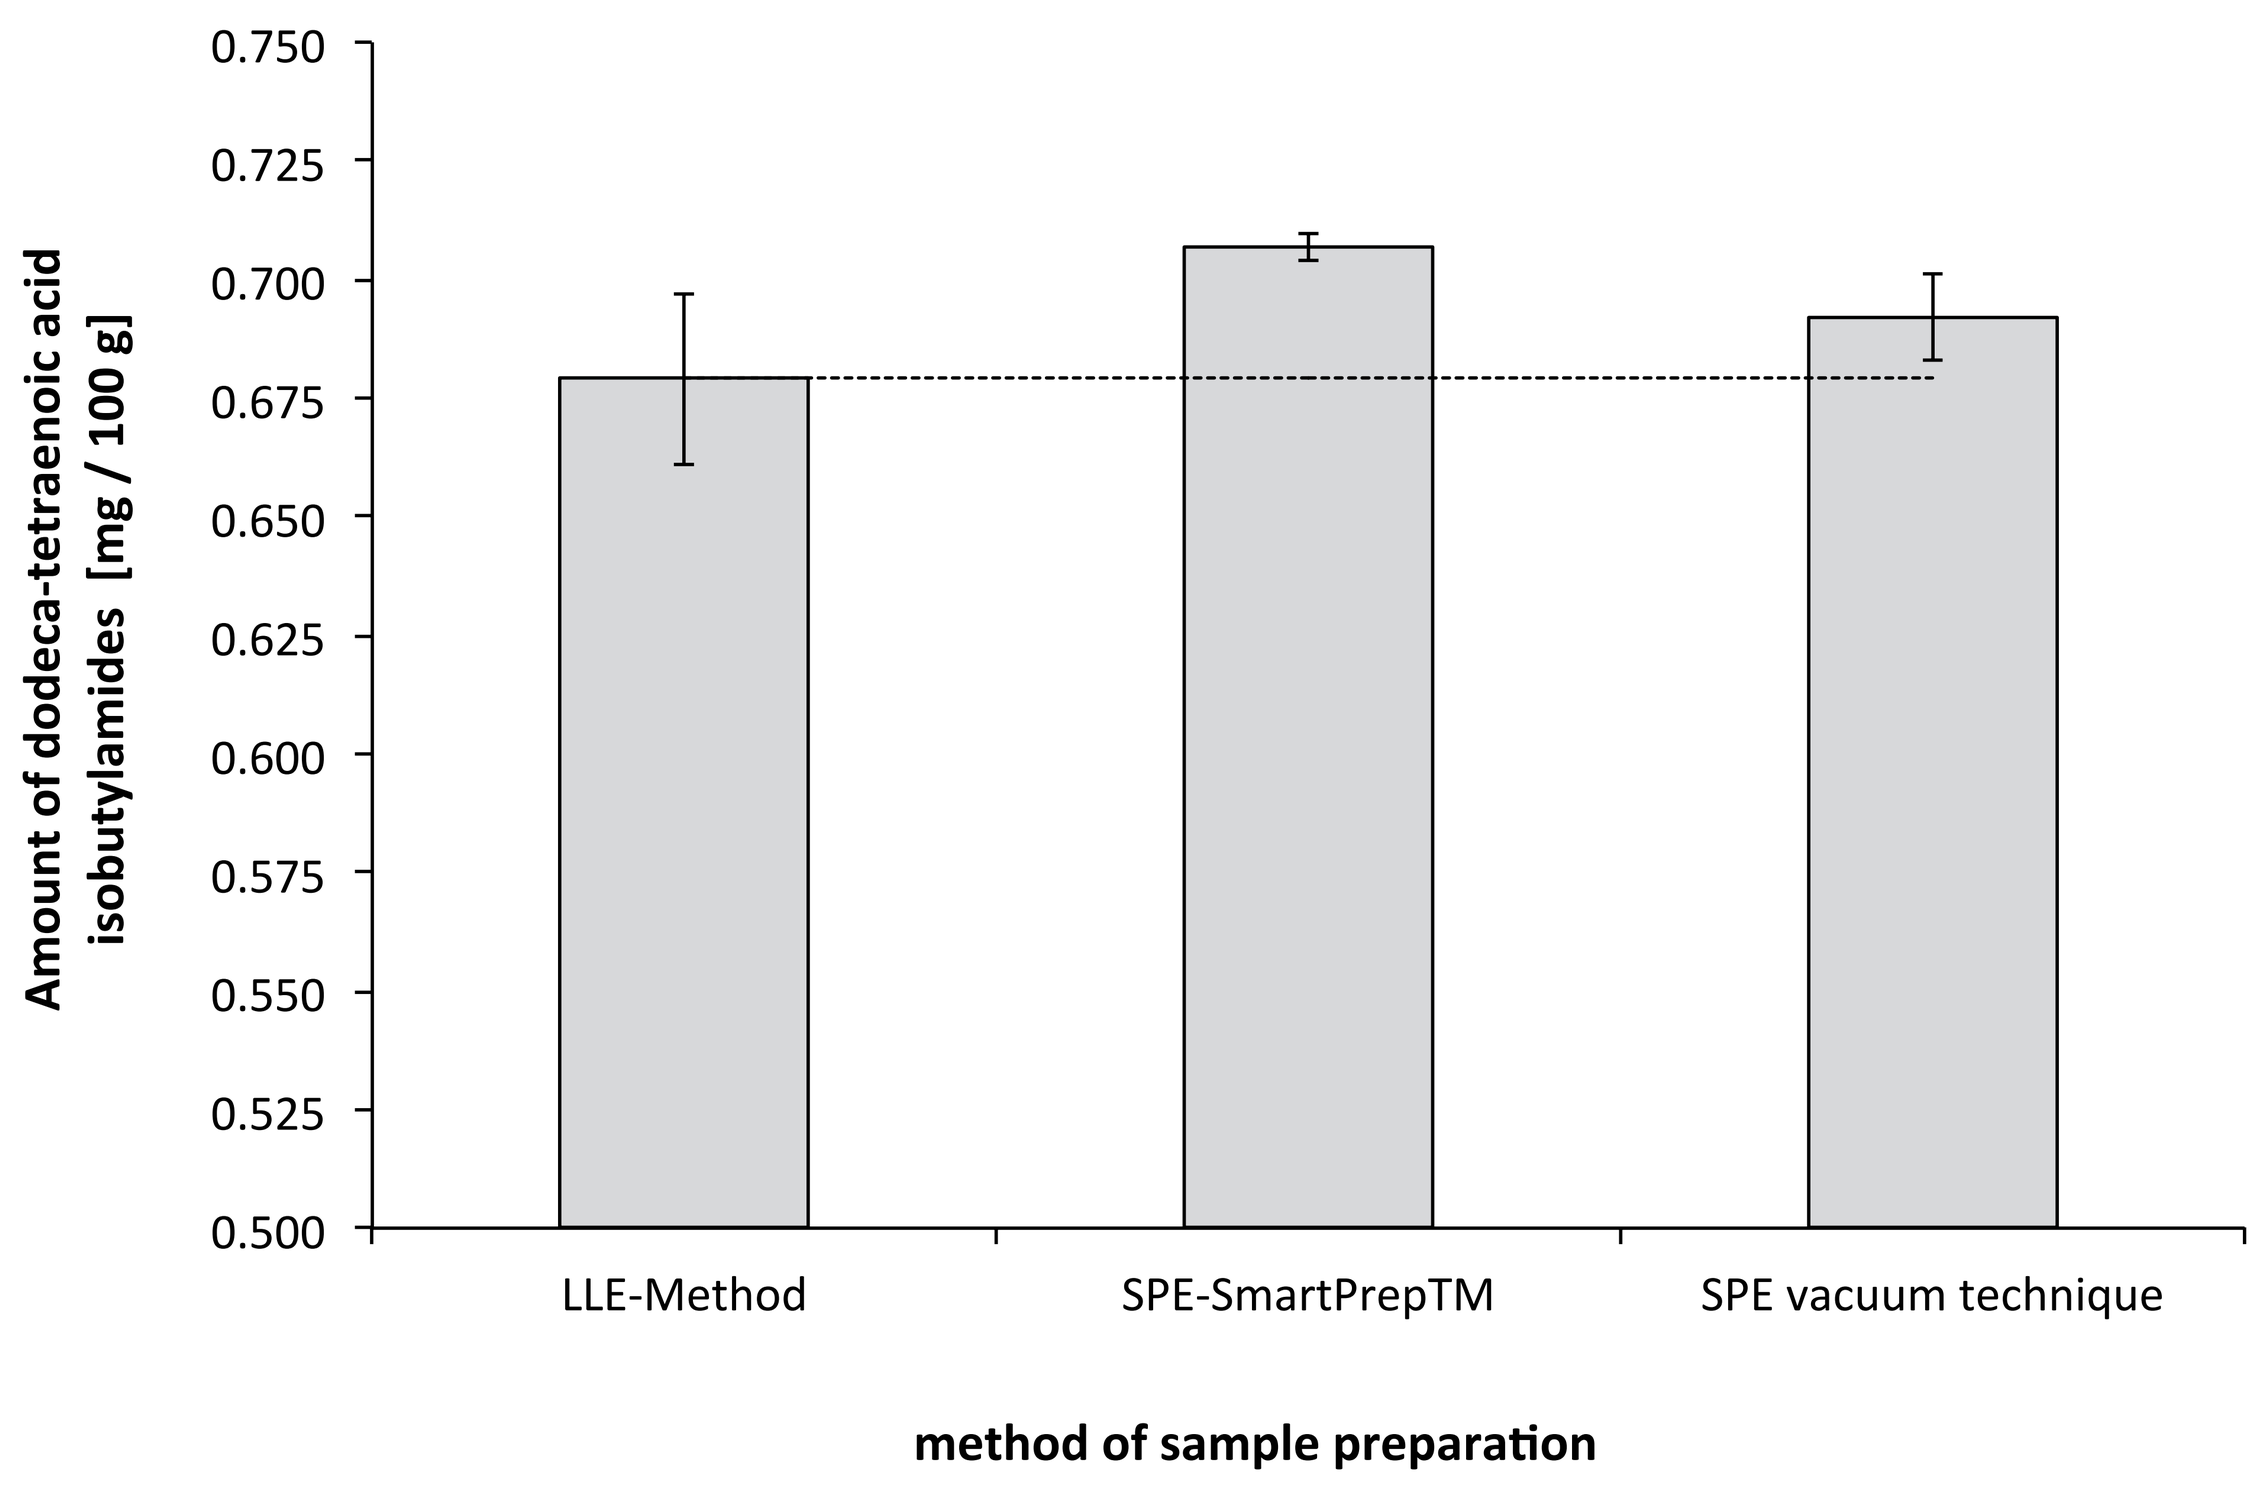
0.750
0.725
0.700
0.675
0.650
0.625
0.600
0.575
0.550
0.525
0.500
Amount of dodeca-tetraenoic acid isobutylamides [mg / 100 g]
LLE-Method
SPE-SmartPrepTM
SPE vacuum technique
method of sample preparation

Figure 3: Comparison of the dodeca-2E,4E,8Z,10E/Z-tetraenoic acid isobutylamide in the Echinacea preparation using LL-Extraction, SmartPrep™ and manual SPE vacuum technique for sample preparation

The displayed chromatograms (A, B and C) in figure 4 show the UPLC chromatograms of dodeca-2E, 4E, 8Z, 10E/Z-tetraenoic acid isobutylamides in the Echinacea preparation using three different sample preparations methods. All 3 chromatograms show the 2 characteristic peaks of the alkylamides at 6.5 min, which are used for the assay.

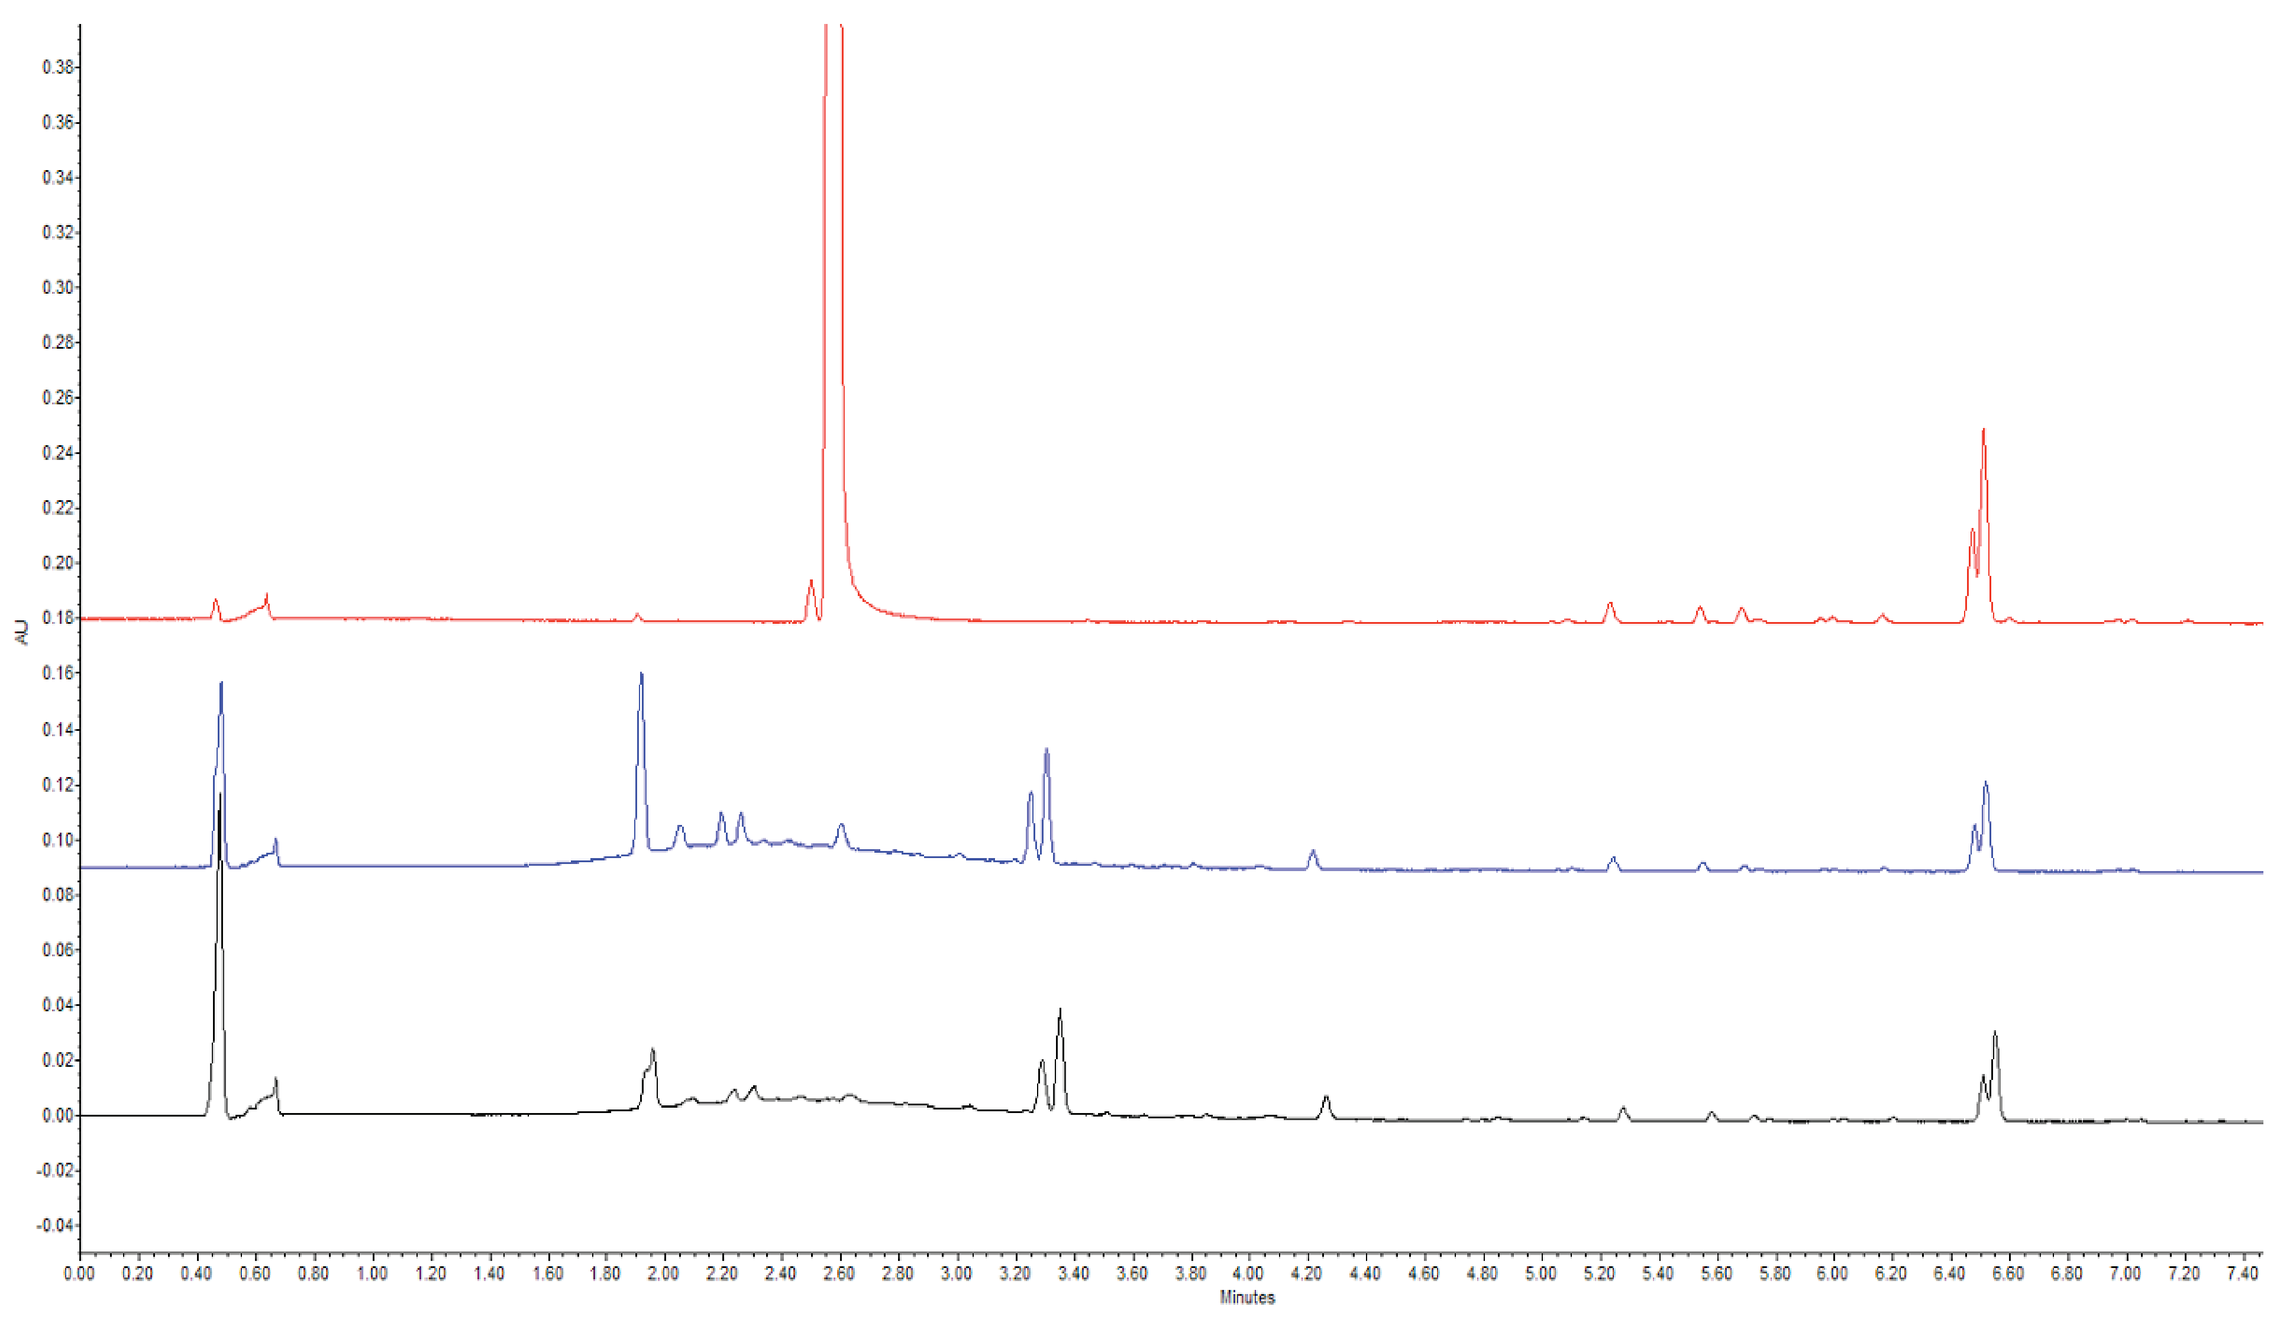
AU
Minutes

Figure 4: Comparison of UHPLC separation of dodeca-2E,4E,8Z,10E/Z-tetraenoic acid isobutylamide (t_R:6.5 min) in Echinacea preparation with the three sample preparation methods. Chromatograms: (A) SmartPrep™, (B) SPE vacuum technique, (C) LLE according to test procedure. Detection at 260 nm.

Discussion

Based on the results of the tests with varying parameters on the automized Solid-Phase-Extraction system a new SPE sample preparation method was developed and validated. The characterization of the SmartPrep™ at various parameters, can provide a basis for further development of methods.

By using the newly established SPE sample preparation on the SmartPrep™, a 4.1% higher yield of dodeca-2E,4E,8Z,10E/Z-tetraenoic acid isobutylamide was achieved, compared to the previously validated LLE method and also a 2.1% higher yield as in case of the manual SPE vacuum technique. This might be due to a completness of extraction from the matrix. The undesirable disturbances in the chromatogram of the Echinacea preparation could be reduced by means of the SPE purification technique, using 3 ml RP-18 cartridges. Herewith, a successful chromatographic separation and detection can be guaranteed. The suitability of the SPE method was documented during validation according to ICH standards; therefore the product specific requirements for content assay of the marker substance are fulfilled.

Conclusion: Automated sample preparation may be a reliable means for extraction of analytical markers from a complex matrix. Careful optimisation is necessary, as there are many parameters to consider.

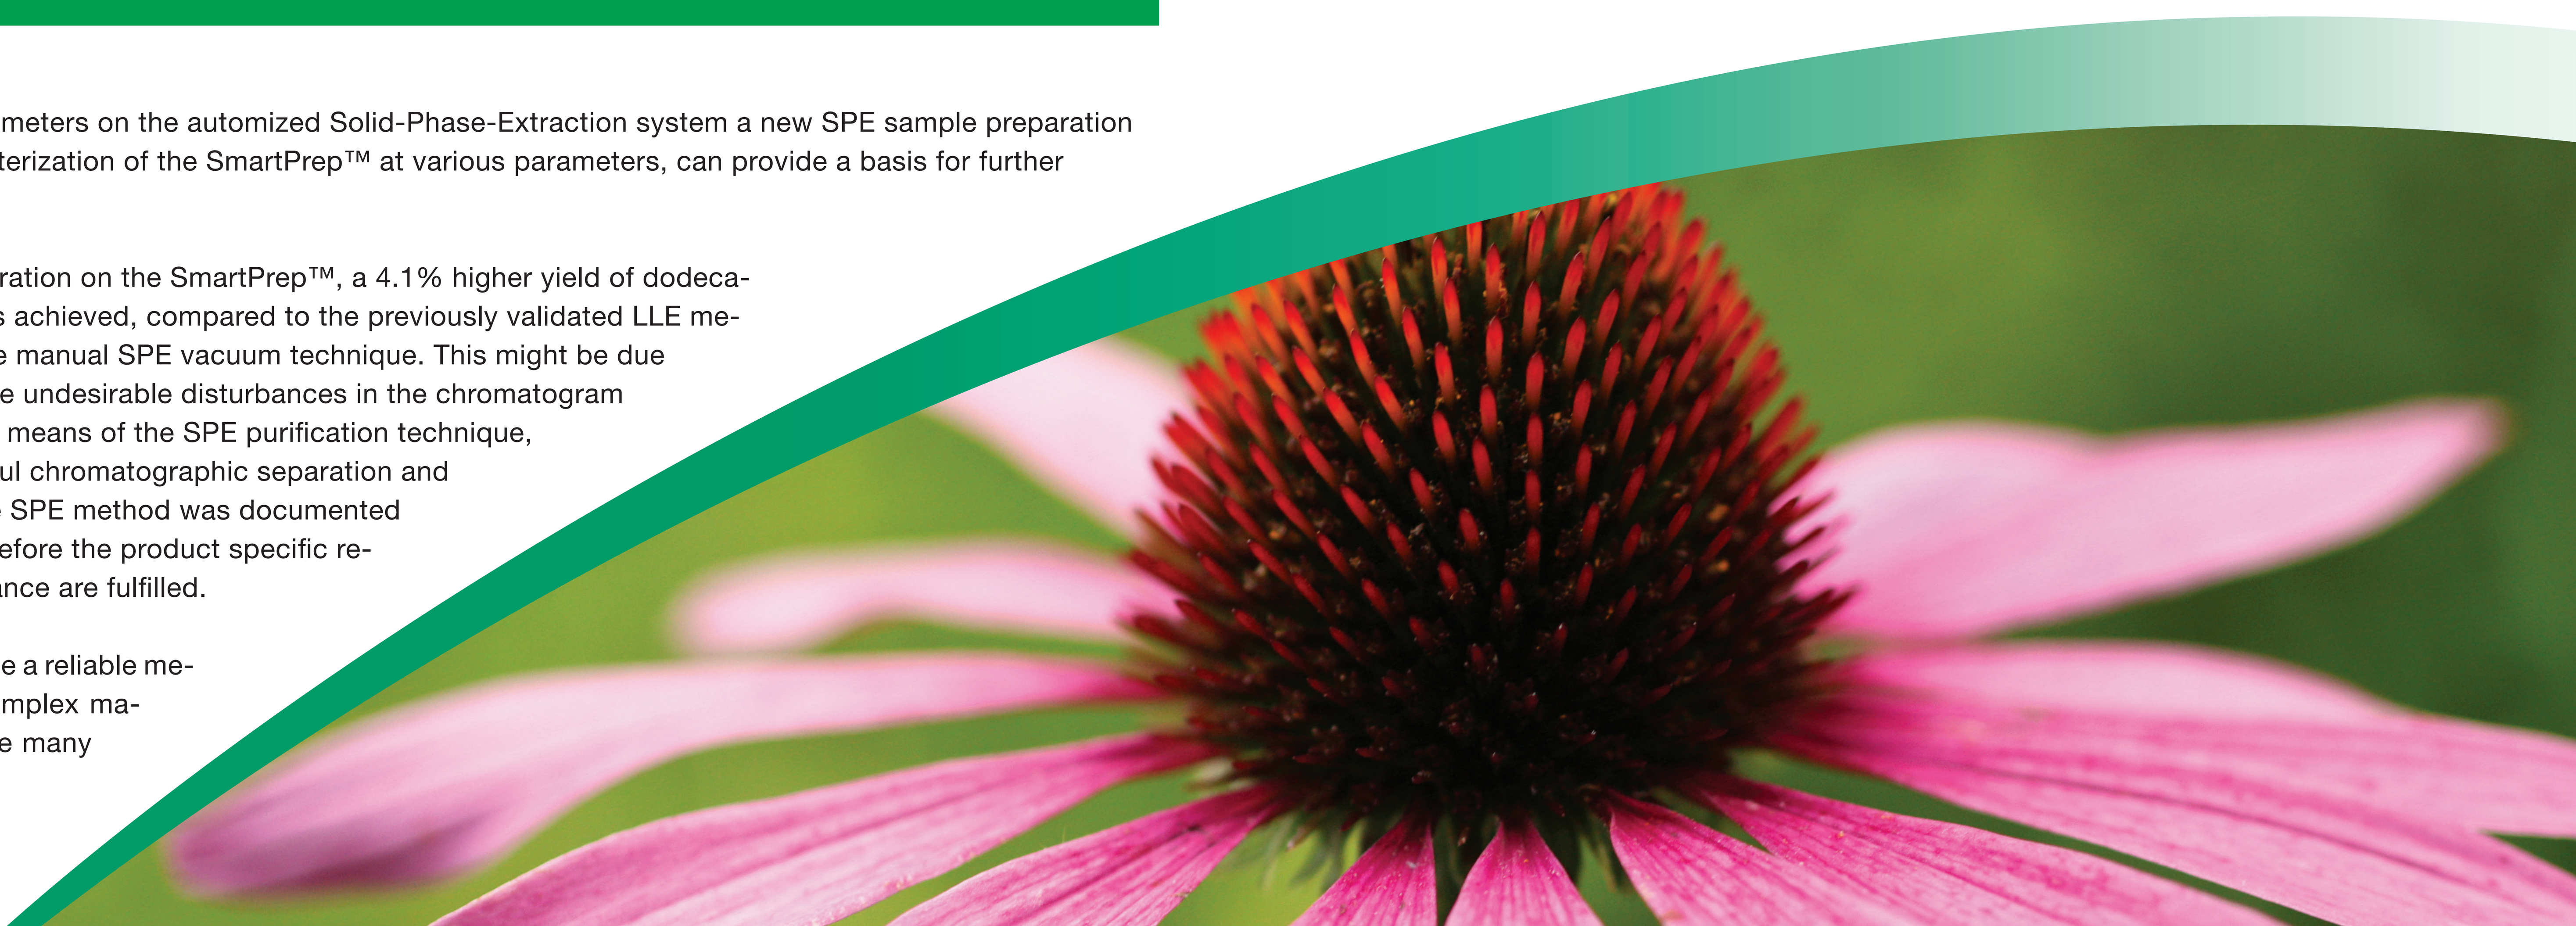