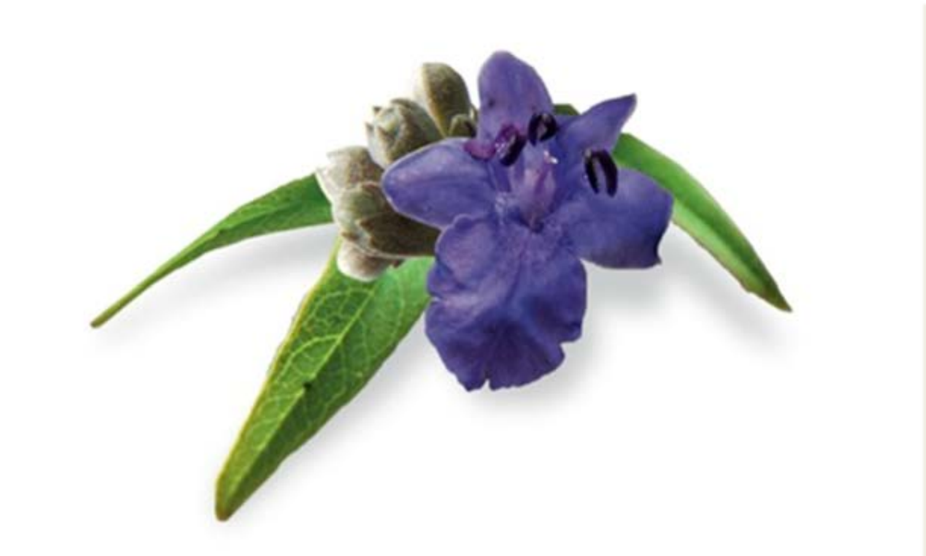

Vitex Agnus-Castus Extract (Ze 440) Improves Symptoms in Women with Menstrual Cycle Irregularities

R. Eltbogen[1], M. Litschgi[2], U. E. Gasser[3], S. Nebel[4] and C. Zahner[4]

[1] Department of Gynecology and Obstetrics, Bürgerspital, Solothurn, Switzerland; [2] Consultant for Gynecology and Obstetrics, Schaffhausen, Switzerland; [3] ClinResearch Ltd., Aesch, Switzerland; [4] Zeller Medical AG, Romanshorn, Switzerland

Ze 440 – Vitex agnus-castus Extrakt

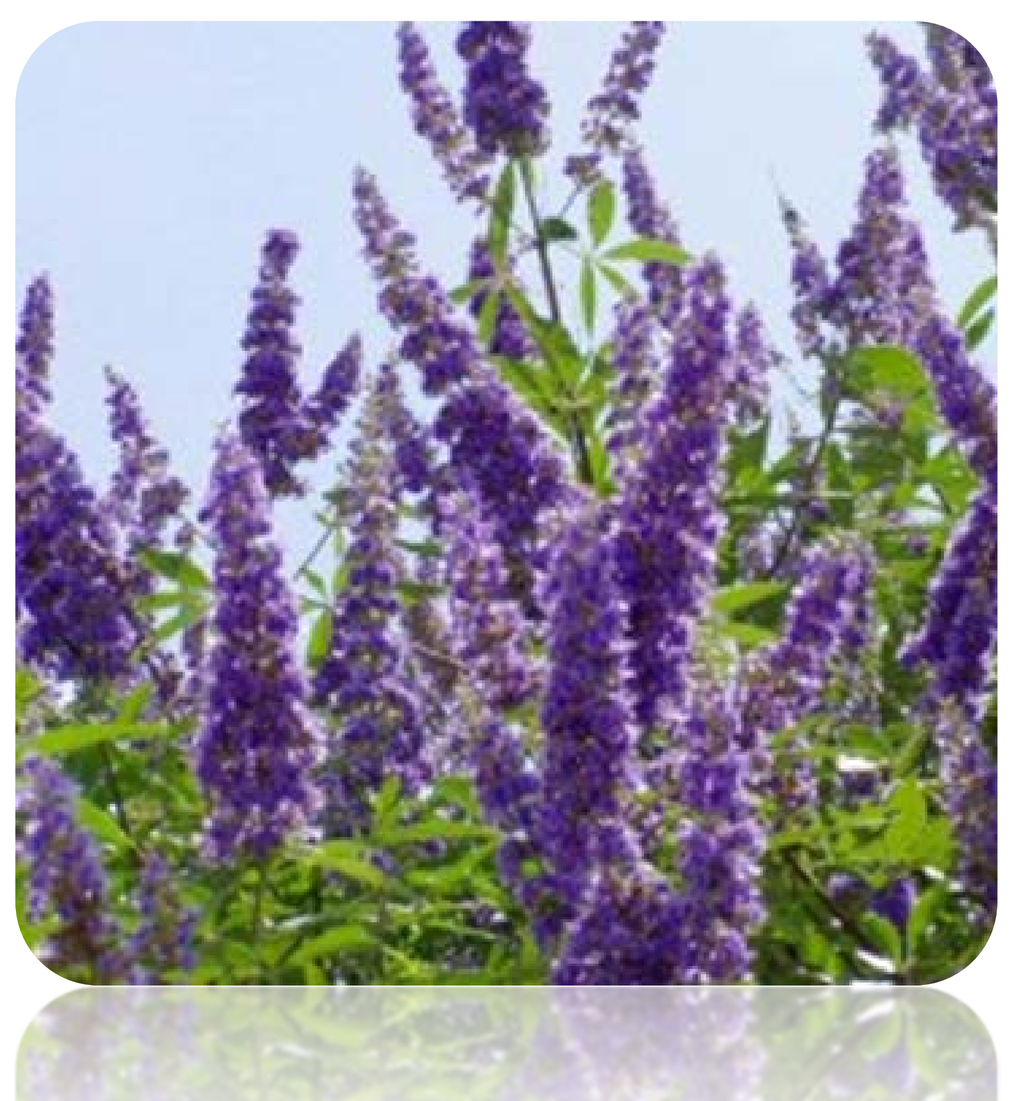

Vitex agnus-castus L.

The fruit of Chasteberry (*Vitex agnus-castus* L.; VAC) has been widely used in Europe for gynaecologic conditions such as the premenstrual syndrome (PMS) and menstrual cycle irregularities (MCIs) including polymenorrhoea, oligo-menorrhoea or amenorrhoea, as well as cyclical breast discomfort and dysfunctional uterine bleeding.

MCIs and PMS are common disorders in women during their reproductive lifespan. The prevalence for MCIs is about 20% in young women and for PMS about 20% to 40% [1].

While the outcomes of recent clinical investigation established the benefit of VAC extracts in PMS, fewer studies were conducted to evaluate the effect on menstrual cycle irregularities (MCIs) and specific symptoms.

Study Aims

- To evaluate the effects of Ze 440 in women suffering from menstrual cycle irregularities (MCIs), such as polymenorrhoea, oligomenorrhoea and amenorrhoea.
- To assess the effect of Ze 440 on other symptoms related to menstrual bleeding, including hypermenorrhoea, menometrorrhagia, inter-menstrual bleeding, ovulation bleeding, and premenstrual and postmenstrual bleeding and dysmenorrhoea.

Study Design

- Non-interventional, observational study conducted in Switzerland
- Treatment with Ze 440 during three subsequent menstrual cycles
- Symptoms associated with MCIs and menstrual bleeding were assessed at baseline visit (BV) and at a follow up visit (FV).

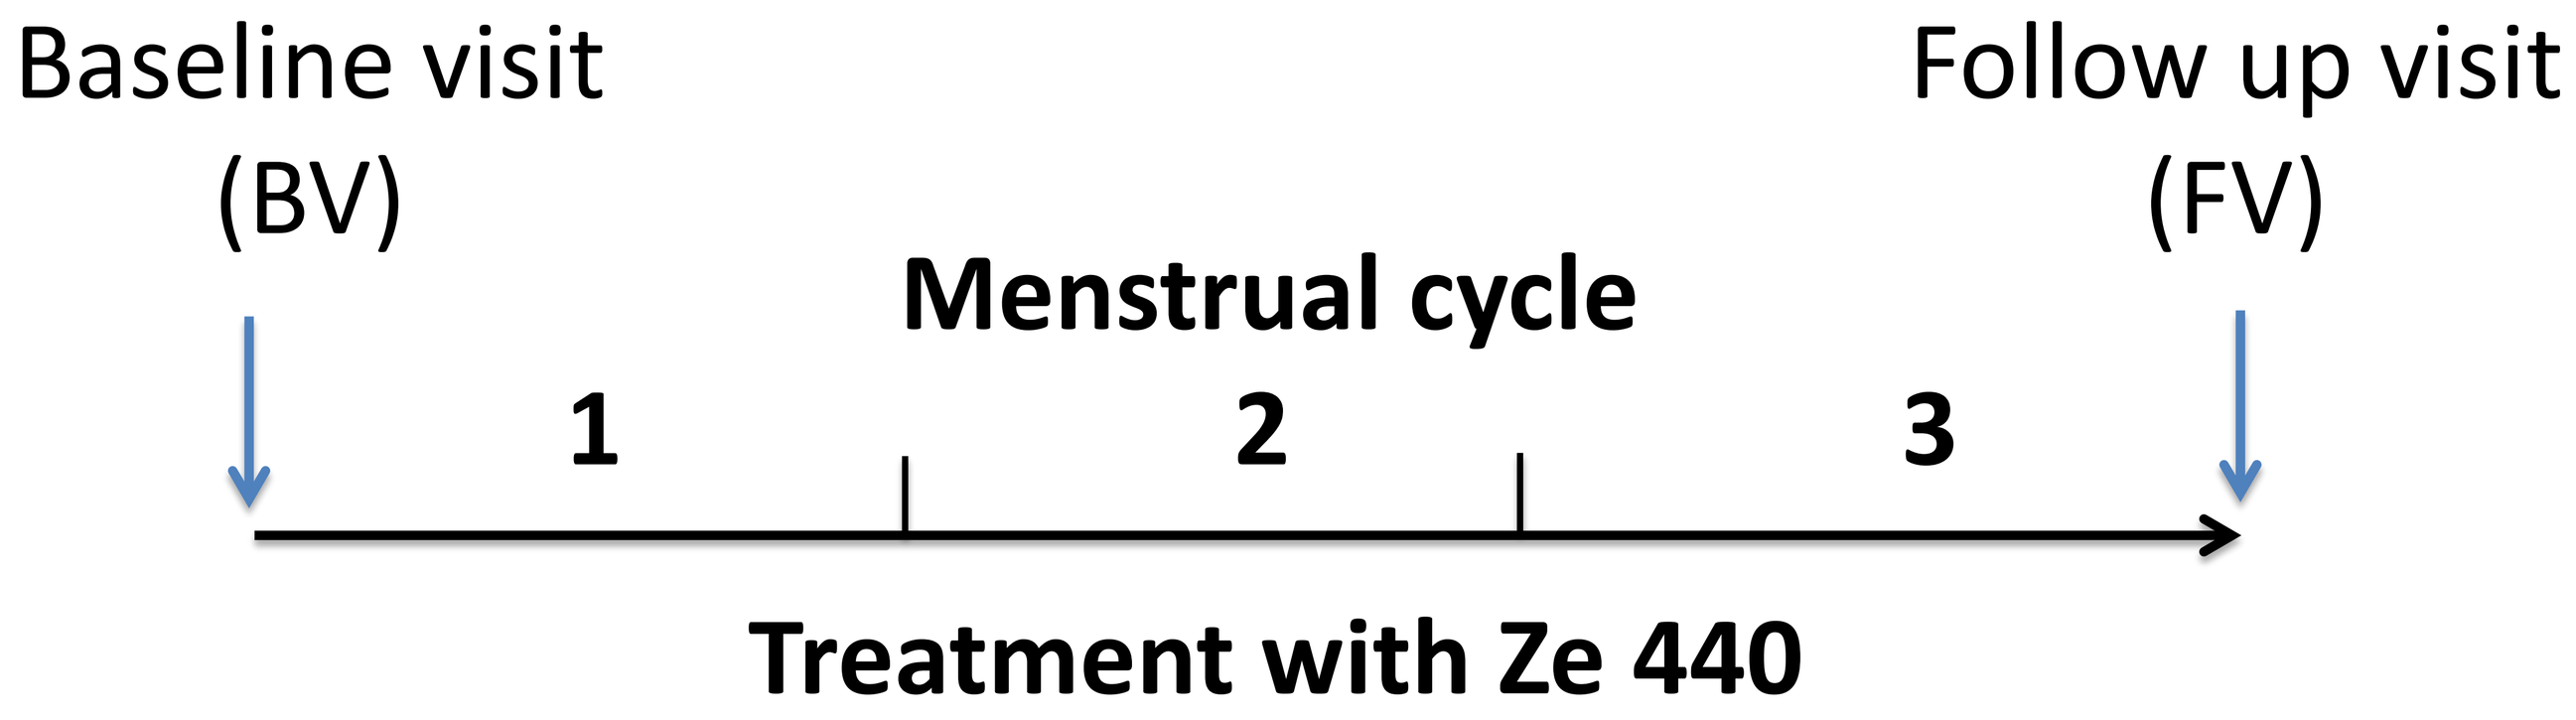
Baseline visit (BV)
Follow up visit (FV)
Menstrual cycle
1
2
3
Treatment with Ze 440

Data collection and analysis

- October 2008 – October 2011
- 43 Swiss physicians (mainly gynecologists) participated
- 211 patients suffering from MCIs were included

Descriptive statistical analysis of all recorded data was performed using SAS (version 9.2). Changes from BV to the FV were analysed.

Results

- Of the 211 patients included, the majority was treated with 20mg Ze 440 daily (recommended dose).
- The proportion of patients with remission or improvement of MCIs (all in all) and specific symptoms such as polymenorrhoea, oligomenorrhoea and amenorrhoea was 79% to 85% at the FV (Fig. A).
- The proportion of patients with remission or improvement of symptoms related to menstrual bleeding was 60% to 88% at FV (Fig. B).

Fig. A) Improvement of MCIs in general and on specific symptoms at FV:

Percent of symptoms
■ resolved, ■ improved,
■ unchanged ■ worsened
■ missing values

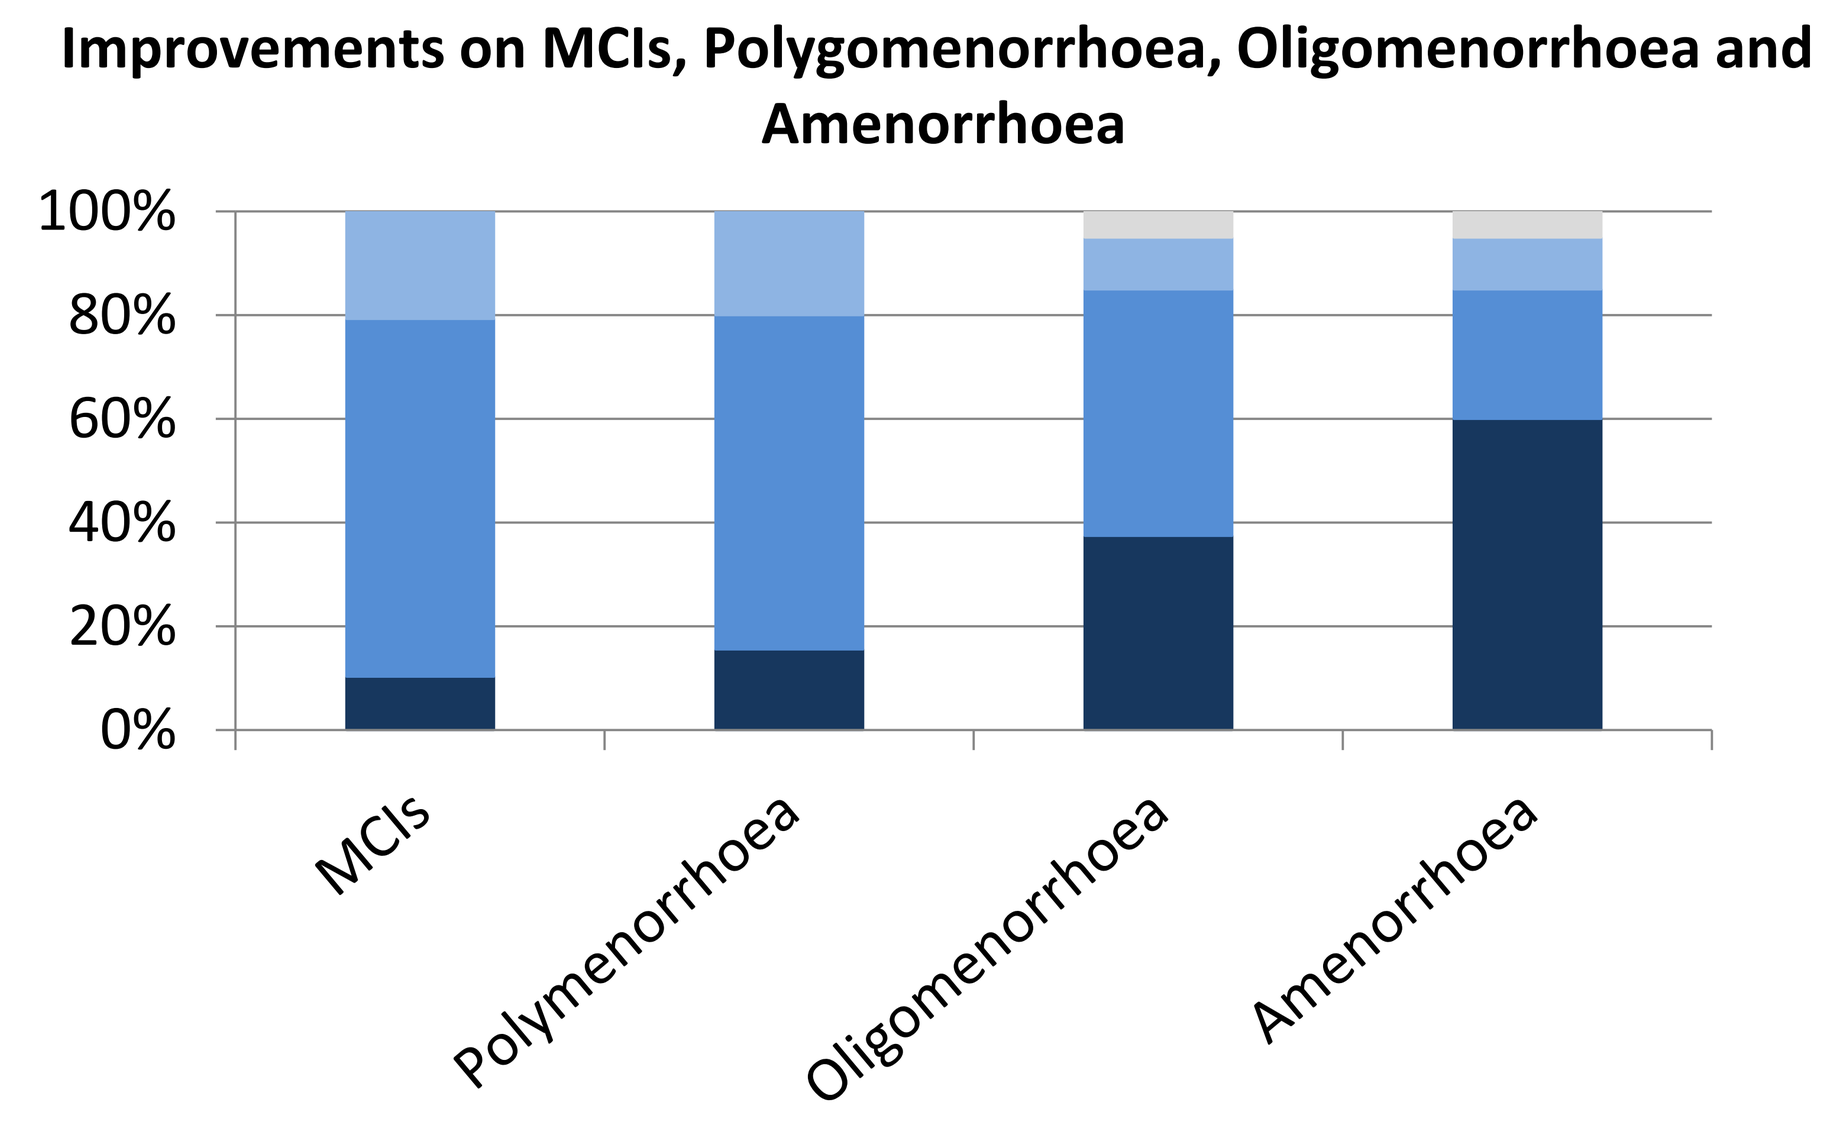
Improvements on MCIs, Polygomenorrhoea, Oligomenorrhoea and Amenorrhoea
100%
80%
60%
40%
20%
0%
MCIs
Polymenorrhoea
Oligomenorrhoea
Amenorrhoea

Fig . B) Improvements on symptoms related to menstrual bleeding at FV:

Percent of symptoms
■ resolved, ■ improved,
■ unchanged or ■ worsened
■ missing values

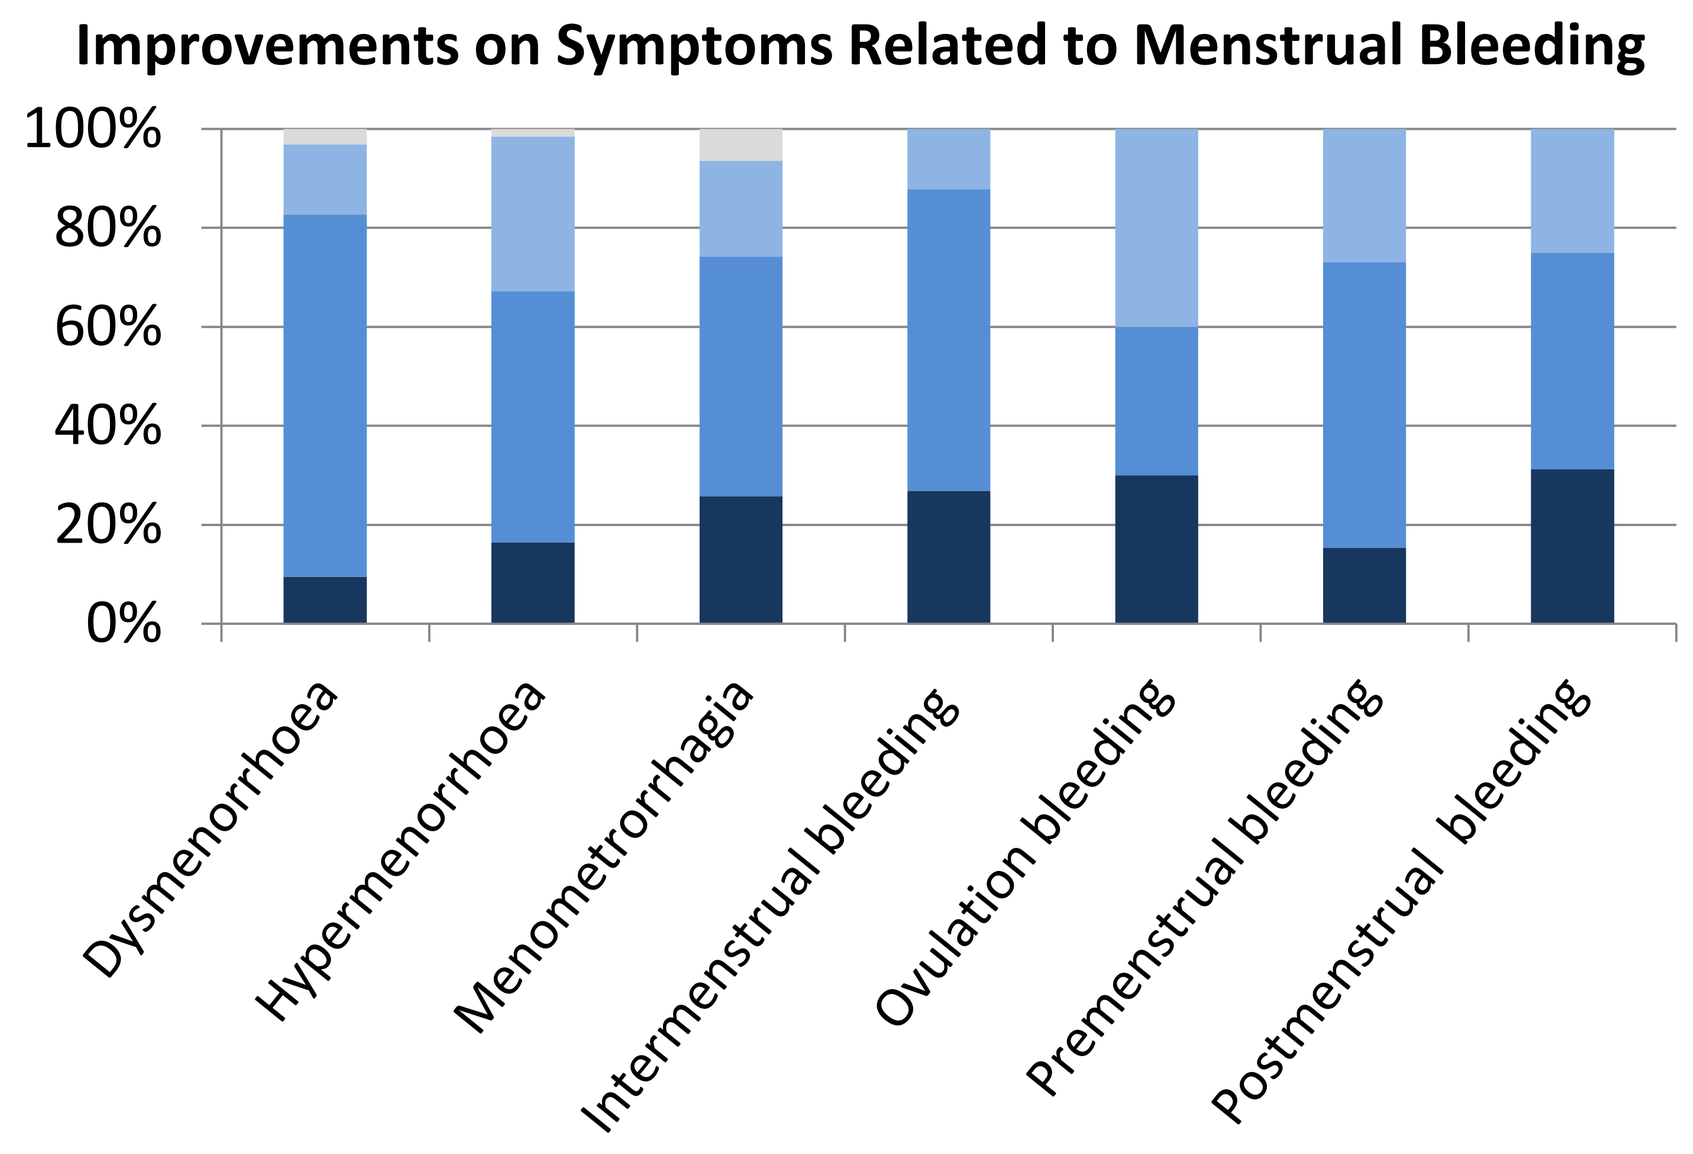
Improvements on Symptoms Related to Menstrual Bleeding
100%
80%
60%
40%
20%
0%
Dysmenorrhoea
Hypermenorrhoea
Menometrorrhagia
Intermenstrual bleeding
Ovulation bleeding
Premenstrual bleeding
Postmenstrual bleeding

- 53 patients (25%) reported an unfulfilled desire to have children at BV. The overall pattern of symptoms in these women at BV was similar compared to the study population; however the rate for MCIs was 80%, and higher as compared overall population. 12 women (23%) got pregnant during the treatment with Ze 440.
- At the FV, 91% of the physicians and 92% of the patients were “satisfied” or “very satisfied” with the achieved treatment outcomes (Fig C.).
- 80% of the patients confirmed their preference to continue the treatment with Ze 440.

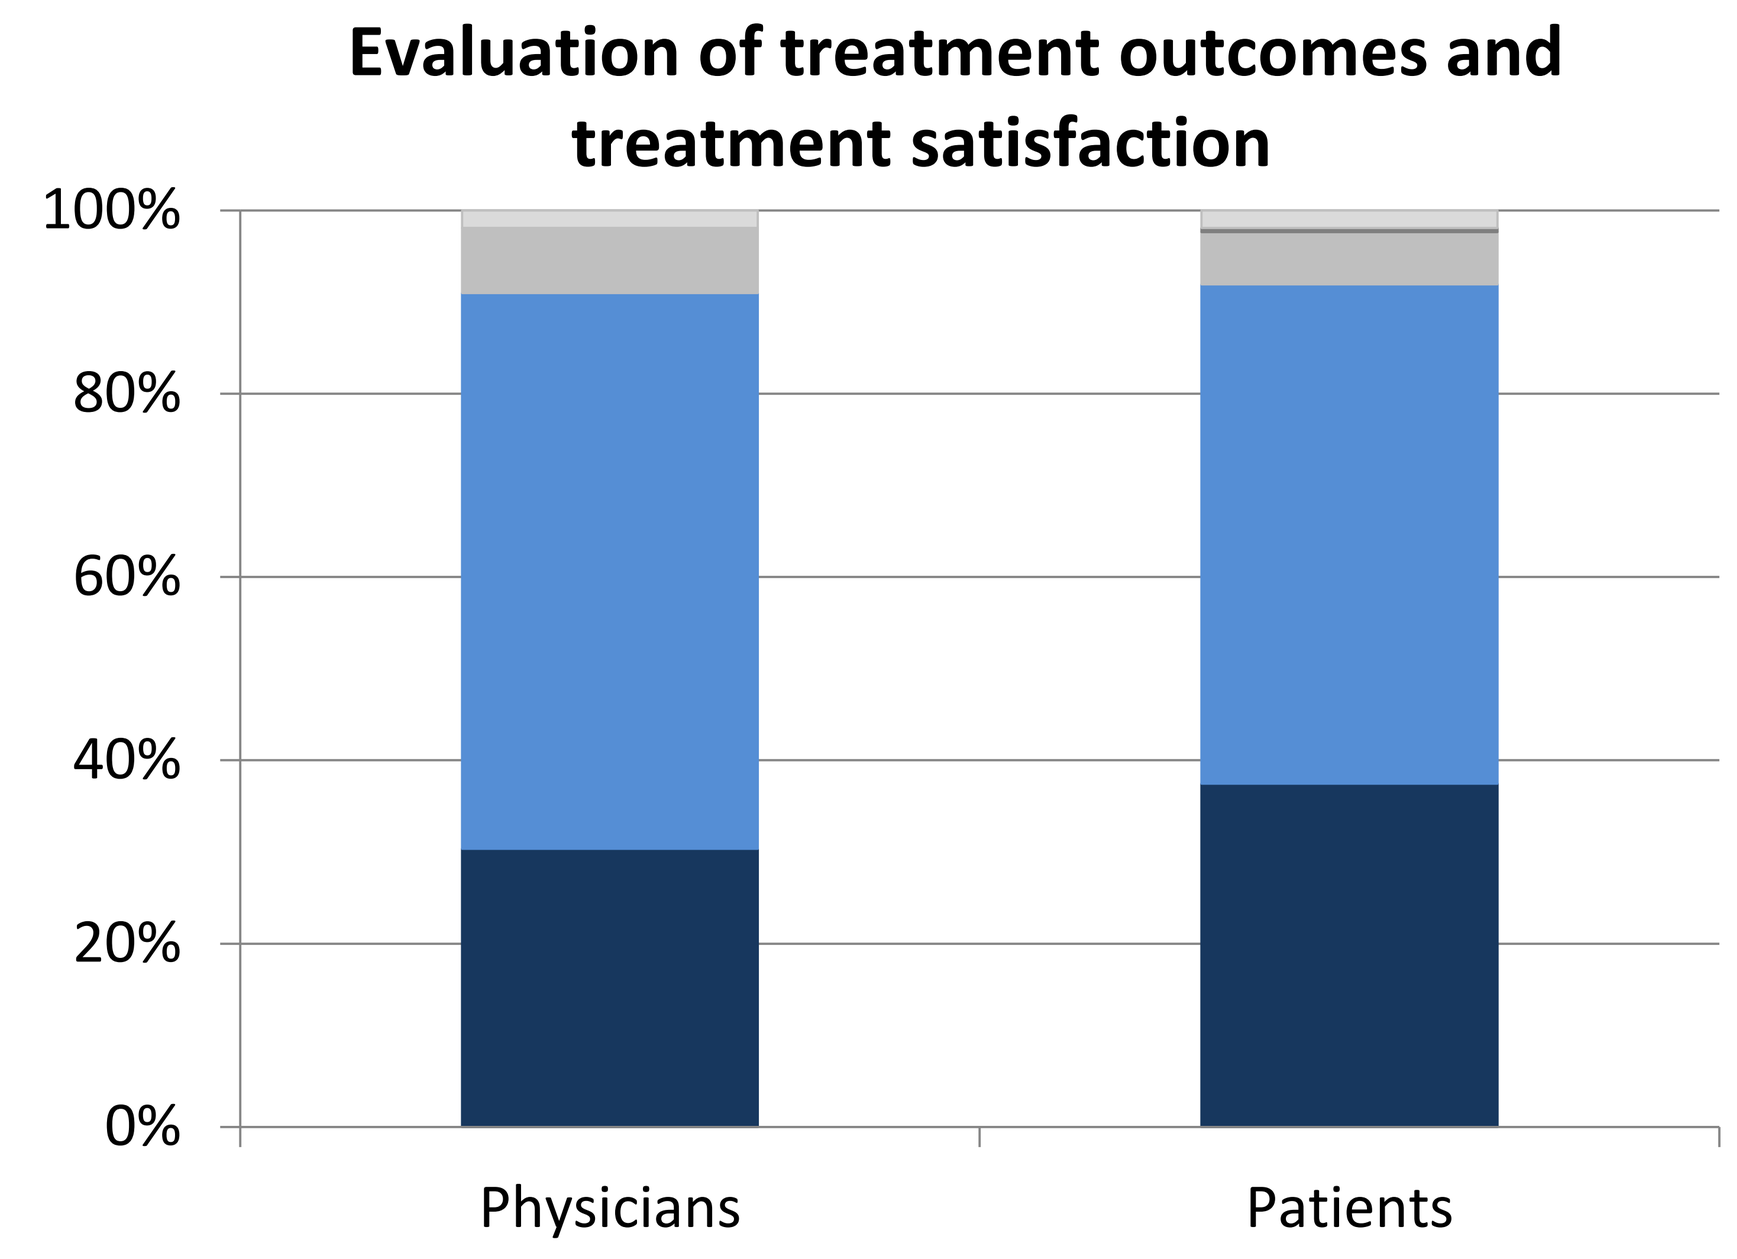
Evaluation of treatment outcomes and treatment satisfaction
100%
80%
60%
40%
20%
0%
Physicians
Patients

Fig. C) Evaluation of treatment outcomes by physicians and treatment satisfaction by patients.

■ very satisfied, ■ satisfied
■ unsatisfied ■ very unsatisfied
■ missing values

Tolerability

Four (2%) women reported an AE (skin rash, erythema, nausea and abdominal pain). All 4 AEs were of mild magnitude and resolved within 4 to 8 weeks.

Conclusion

- ➢ **The observational study in routine medical care showed that treatment with Ze 440 clearly ameliorates MCIs, such as polymenorrhoea, oligomenorrhoea or amenorrhoea.**
- ➢ **Treatment with Ze 440 was well tolerated.**
- ➢ **Treatment with Ze 440 was continued in 80% of the women after the initial treatment during three menstrual cycles.**

Reference:
[1] Tschudin S, Coda Bertea P, Zemp E. Prevalence and predictors of premenstrual syndrome and premenstrual dysphoric disorder in a population-based sample. Arch Womens Ment Health 2010;13:485–494